Extracellular potassium may potentiate the action of a multi-component medication, SKT, in skeletal muscle.

C. Sam[1], D. Terrar[1], P. Noble[2], K. Tasaki[2], D. Noble[2]

[1]Department of Pharmacology, and [2]Department of Physiology, Anatomy and Genetics,
University of Oxford, Oxford, United Kingdom

INTRODUCTION

An accompanying poster (Sam et al 2015) on Shakuyaku-kanzo-to (SKT) showed that in normal physiological conditions the threshold for its action in inhibiting contraction in skeletal muscle is relatively high (1 mg/ml) compared to likely therapeutic levels. What could explain how much lower levels can achieve inhibition and why does it not inhibit general skeletal musculature?

METHODS

The skeletal muscle experiments were performed on guinea-pig and rat phrenic nerve-diaphragm preparation using phrenic nerve stimulation and direct muscle stimulation. The guinea-pigs were Duncan Hartley, 300-500g. Rats were male Long Evans or Wistar, 200-500g. Both were stunned by cervical dislocation.

RESULTS

We determined whether interstitial potassium could be involved since experiments on humans show that interstitial potassium rises substantially during exercise (Green et al, 2000). Increasing extracellular potassium from 5.4 mM to 10.8 mM itself achieves inhibition of muscle contraction which develops with a time course remarkably similar to that produced by SKT. When SKT is then also applied further inhibition of contraction occurs (Figs 1 and 2), but with a much lower threshold concentration, around 0.1 to 0.2 mg/ml.

Both interventions might act on a common component of the relevant cell networks to potentiate each other. Further experiments are required to investigate these and other possible explanations.

We used the Shorten et al (2007) model to determine whether interstitial [K] changes similar to those recorded experimentally can be predicted. The model was coded in CellML format to enable it to run in OpenCOR (http://www.opencor.ws/). Figure 2 shows that even modest frequencies of stimulation of contraction can achieve a significant rise in interstitial potassium. This model is now being developed to enable hypotheses to be explored that might help to explain the synergy between potassium and SKT

FIGURE LEGENDS

Figures 1 and 2. Action of 1 mg/ml SKT on response of rat phrenic nerve-diaphragm showing reduction of twitch in response to both nerve stimulation and direct muscle stimulation remains in the presence of a 100% increase in potassium concentration from 5.4 to 10.8 mM. Vertical bars show standard error. N = 9.

Figure 3. Effect of 1 second tetanus during 20 Hz stimulation on interstitial potassium in the Shorten et al skeletal muscle model. Top trace: computed membrane voltage. Middle : interstitial potassium rises by around 0.4 mM. Bottom: cytosol calcium. Only these 3 key parameters are plotted here, but the model represents many more.

CONCLUSION

Powerful facilitation of the action of SKT by interstitial potassium explains why the medication can relax cramped muscle without dangerous side effects on respiratory or postural muscles. The results are important in showing that SKT is a targetted medication in acting precisely and only on the region affected by the pathological condition. They also contribute to assessing the safety profile.

Acknowledgements and references
This research is conducted by the University of Oxford Innovative Systems Biology Project Sponsored by Tsumura. We thank TSUMURA & CO for their support and for the provision of chemicals.
Green S, Langberg H, Skovgaard D, Bülow J & Kjaer M. (2000). Interstitial and arterial–venous [K+] in human calf muscle during dynamic exercise: effect of ischaemia and relation to muscle pain *Journal of Physiology* **529,** 849-861.
Shorten PR, O'Callaghan P, Davidson JB & Soboleva TK. (2007). A mathematical model of fatigue in skeletal muscle force contraction *Journal of Muscle Research and Cell Motility* **28,** 293-313.
Sam et al (2105) Actions of a multi-component medication, SKT, on skeletal, smooth and cardiac muscle, Physiological Society abstracts.

Figure 1

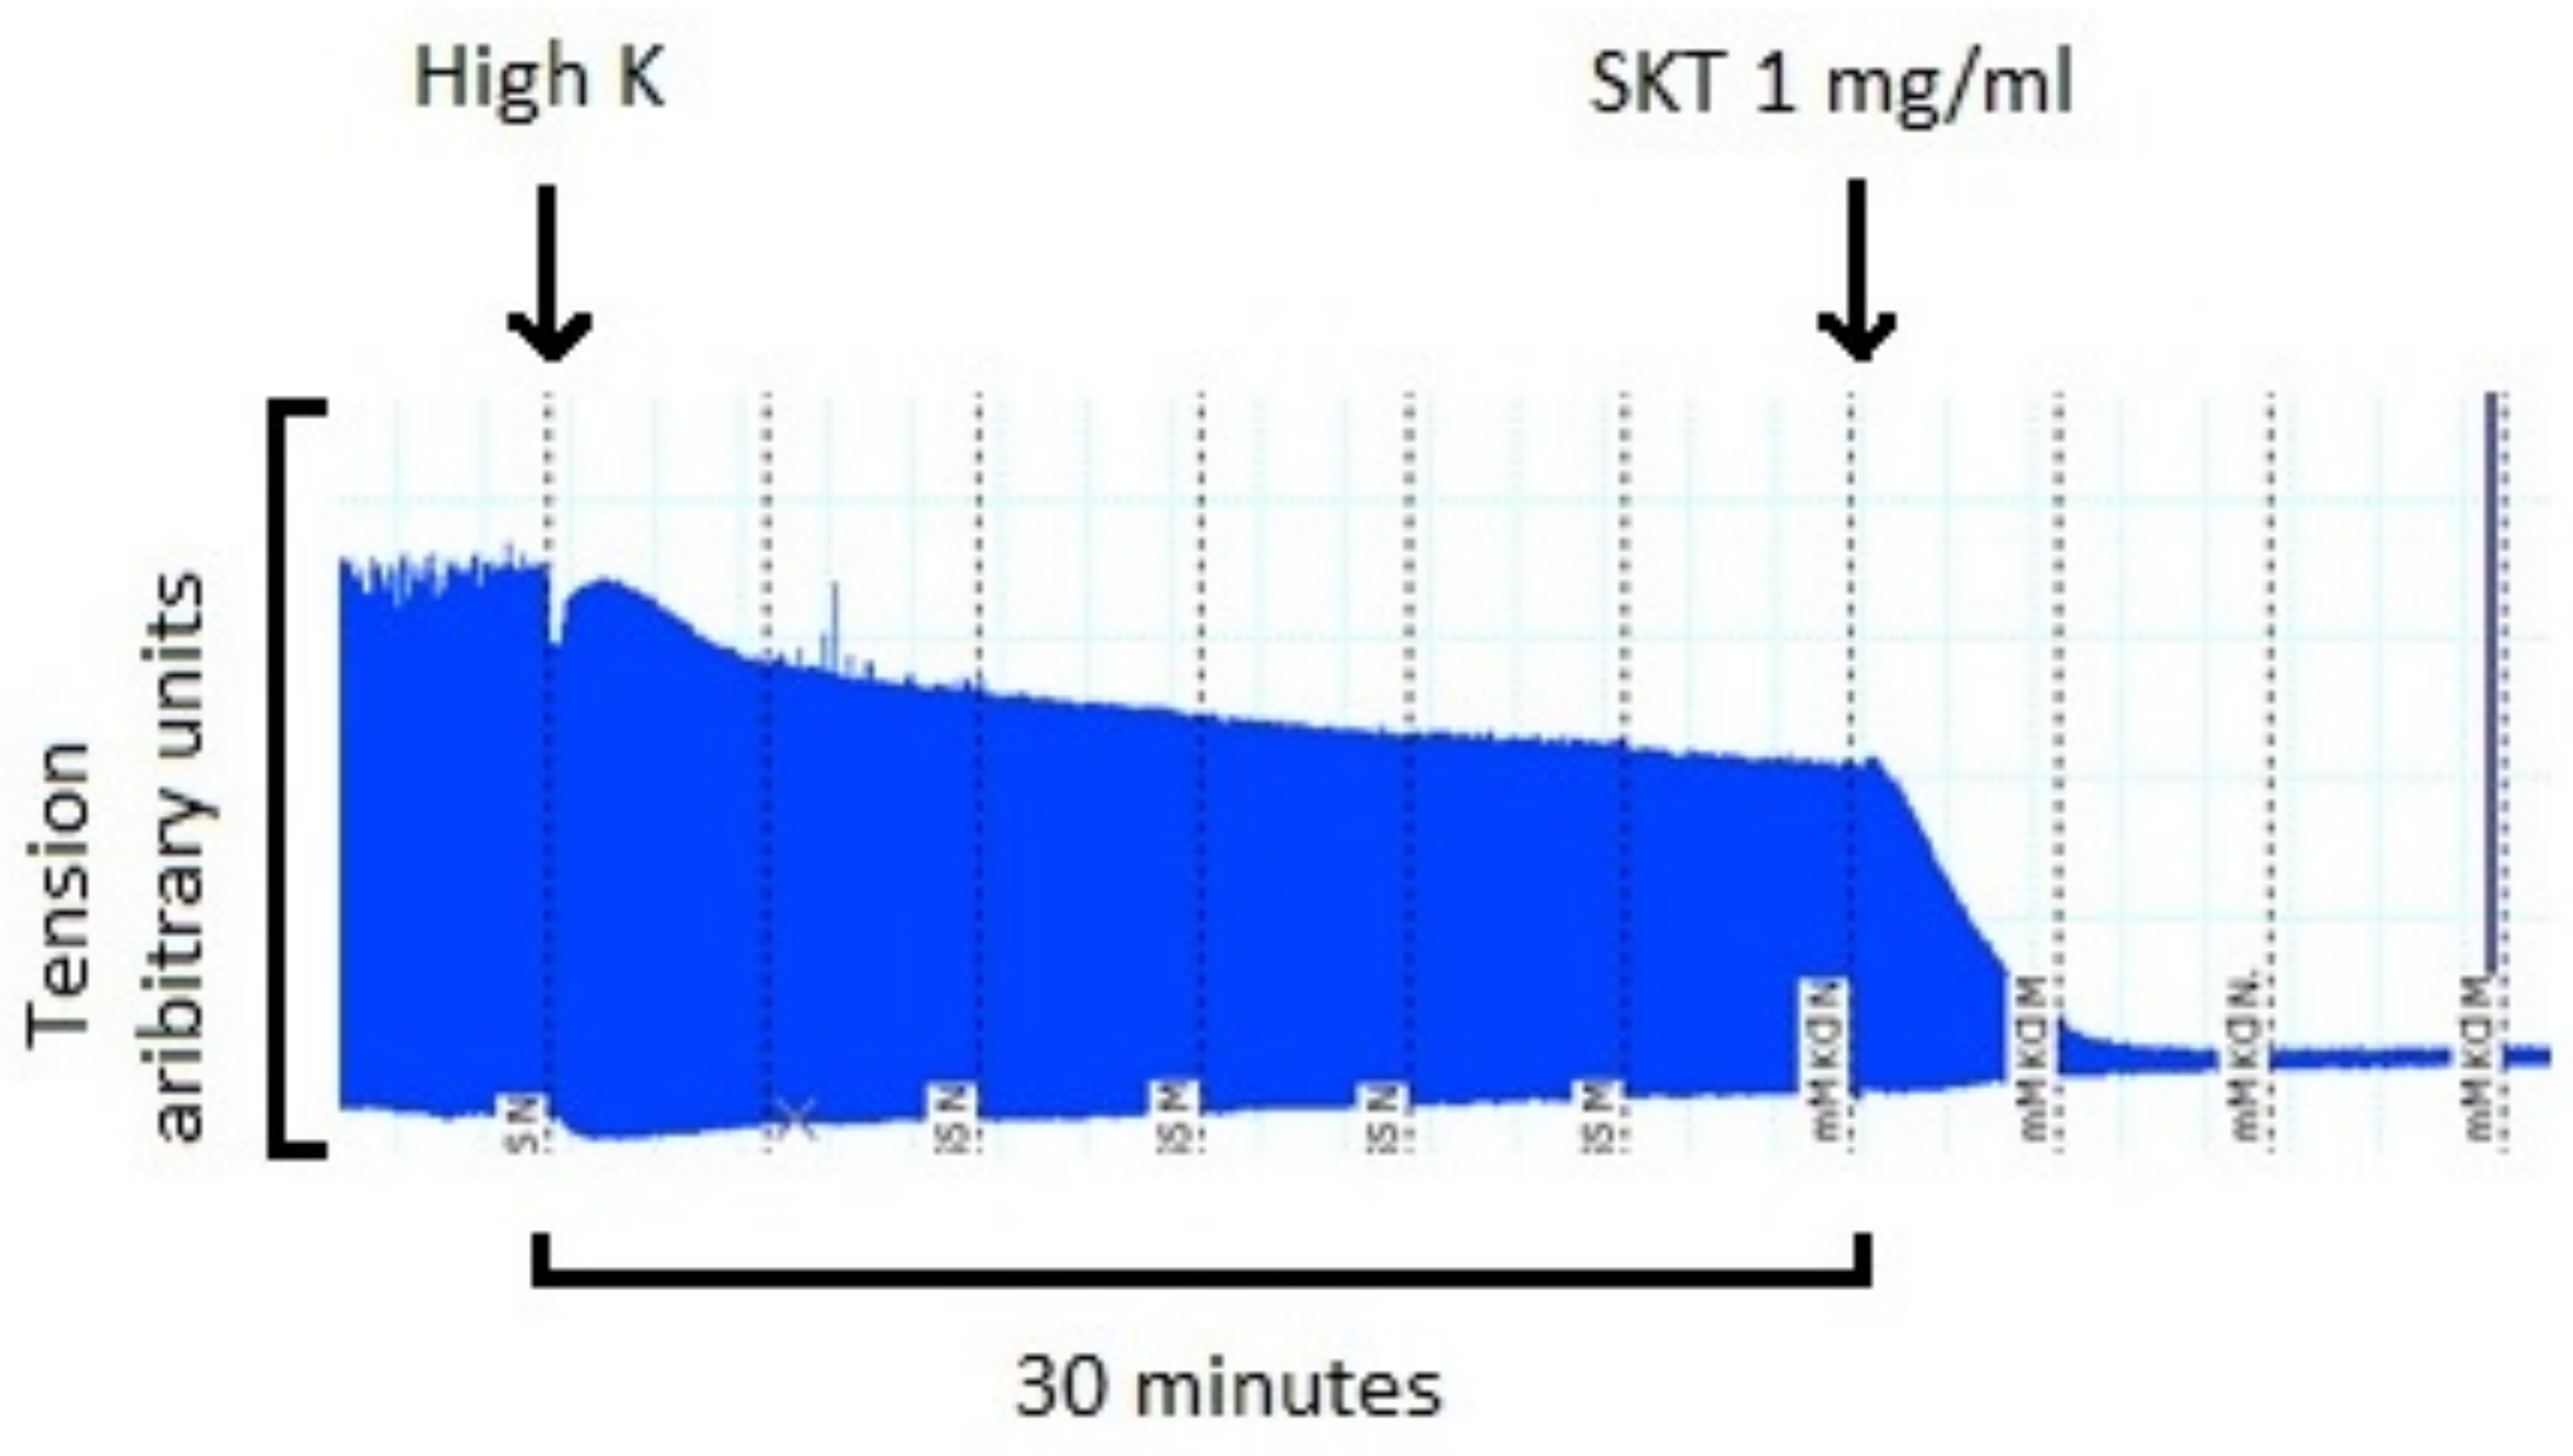
High K
SKT 1 mg/ml
Tension
aribitrary units
30 minutes

Figure 2

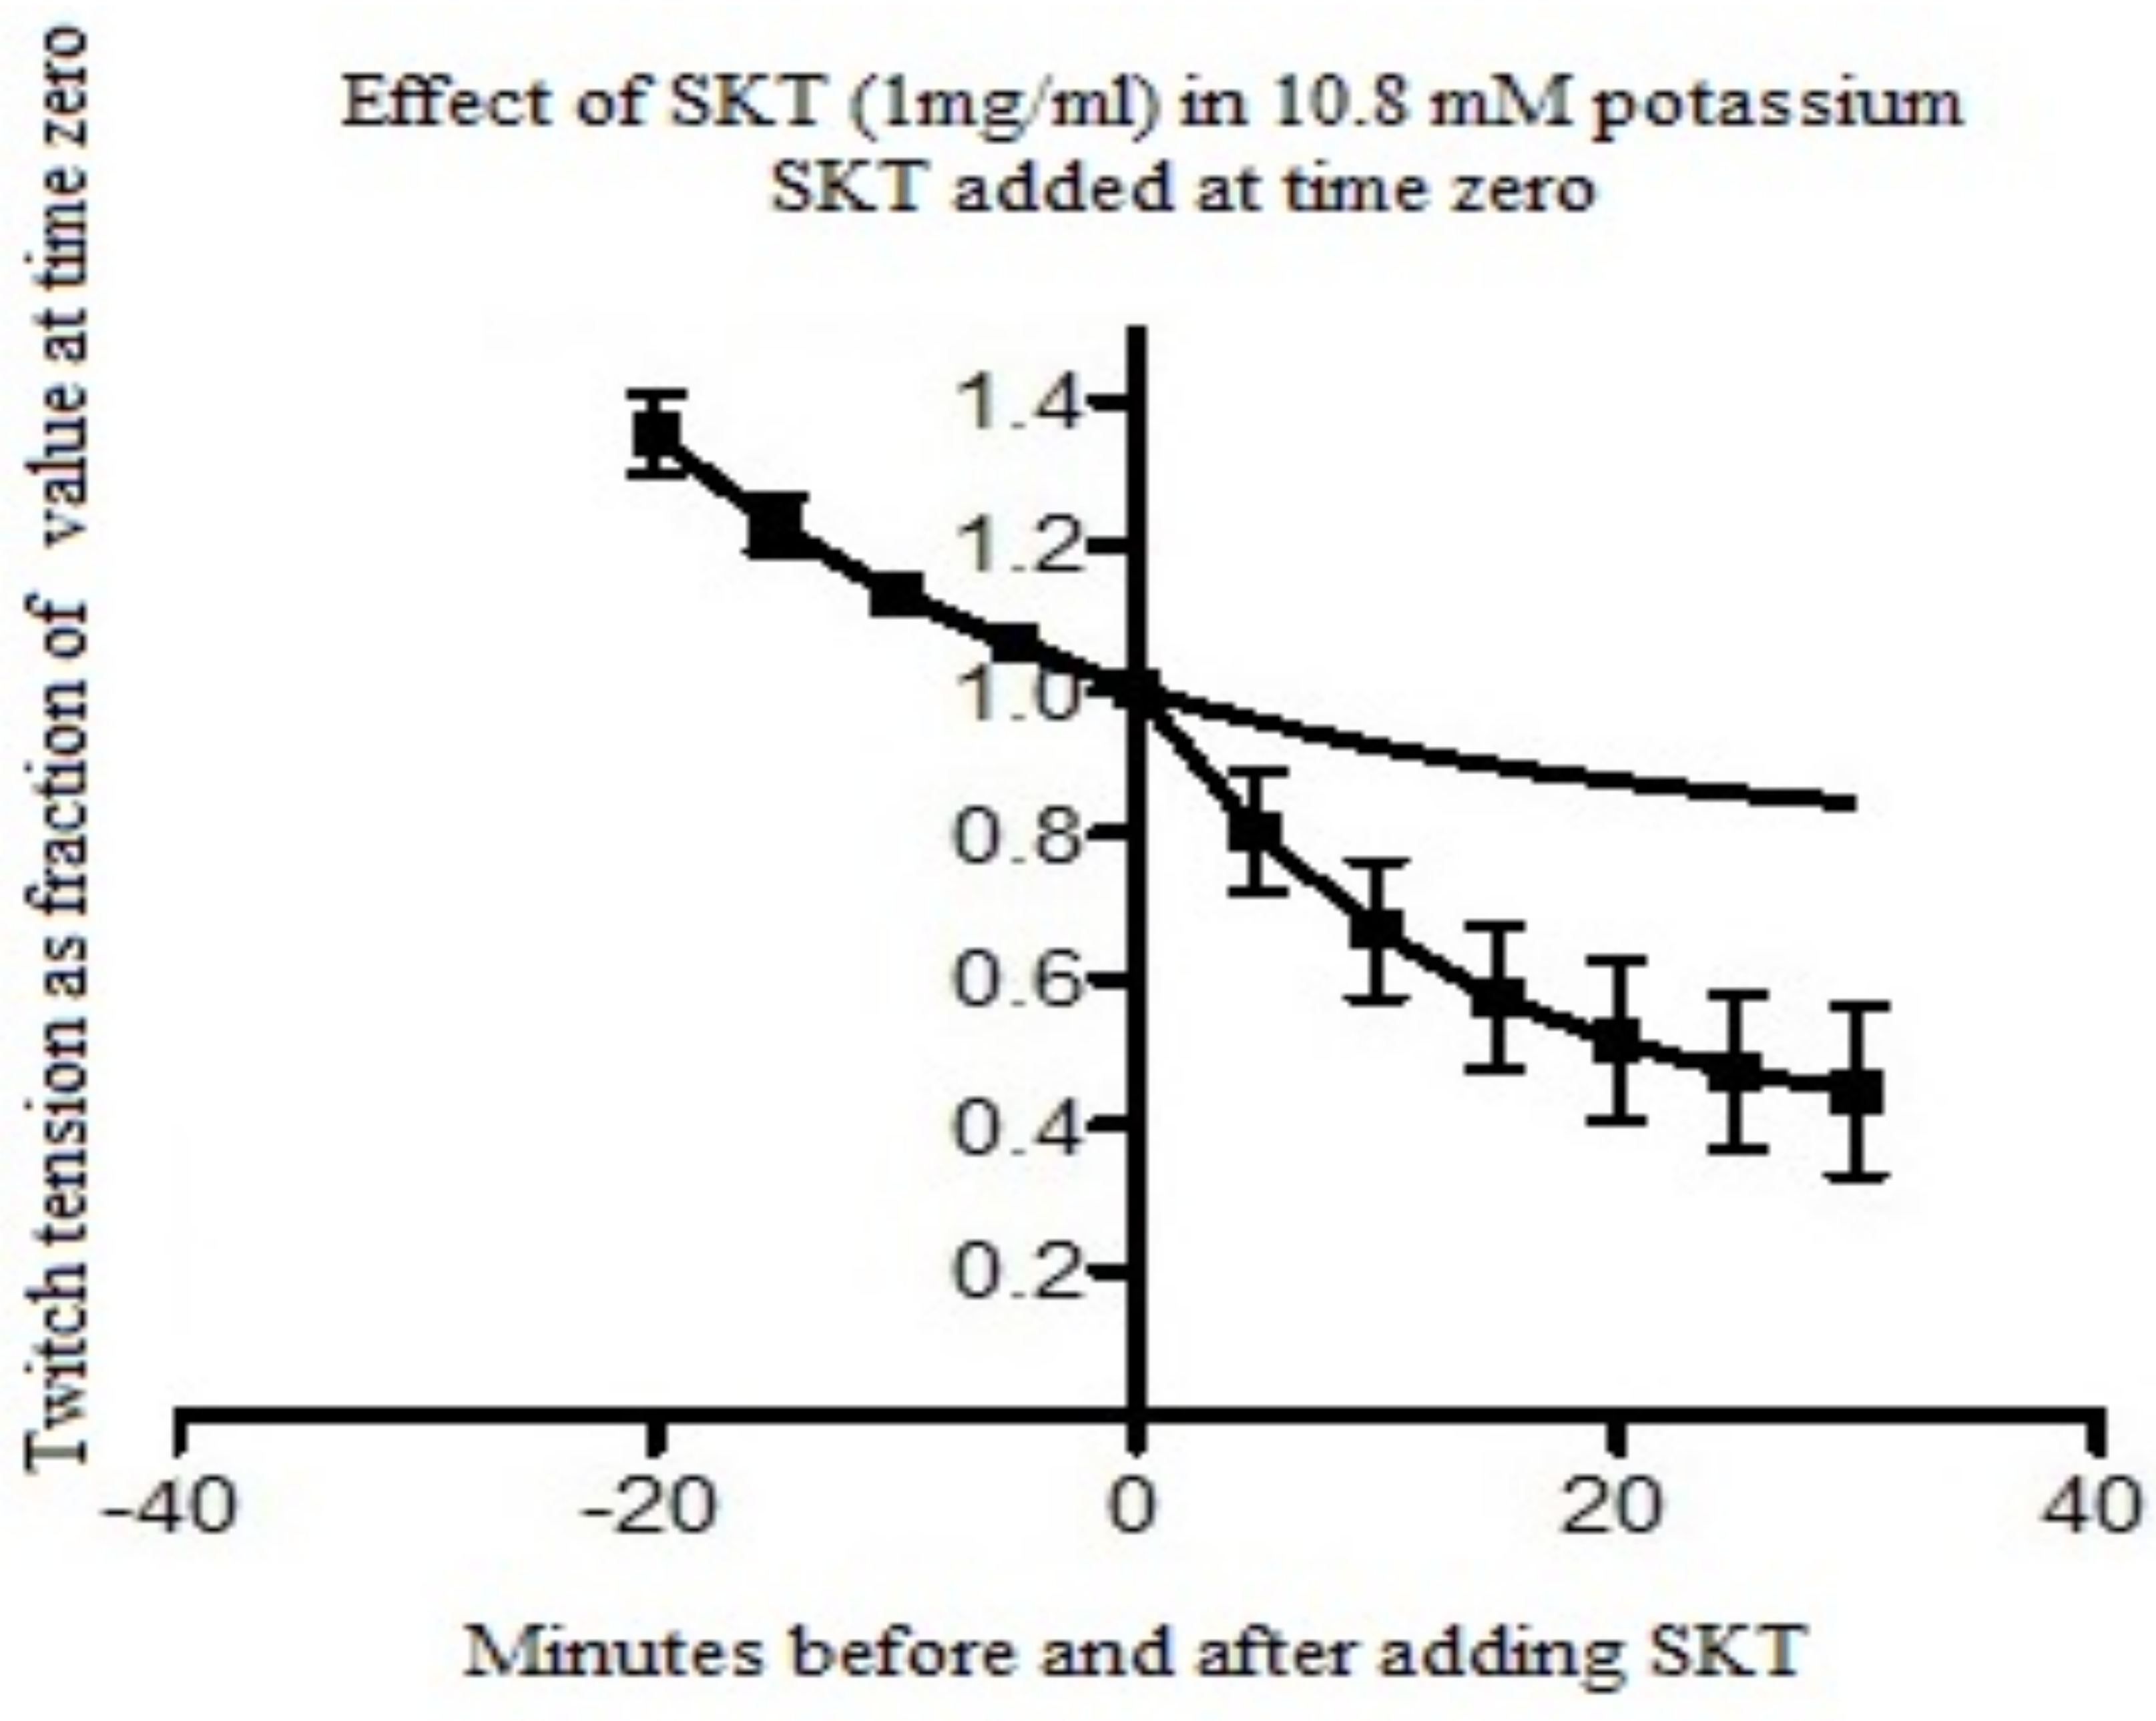
Effect of SKT (1mg/ml) in 10.8 mM potassium
SKT added at time zero
Twitch tension as fraction of value at time zero
1.4
1.2
1.0
0.8
0.6
0.4
0.2
-40
-20
0
20
40
Minutes before and after adding SKT

Figure 3

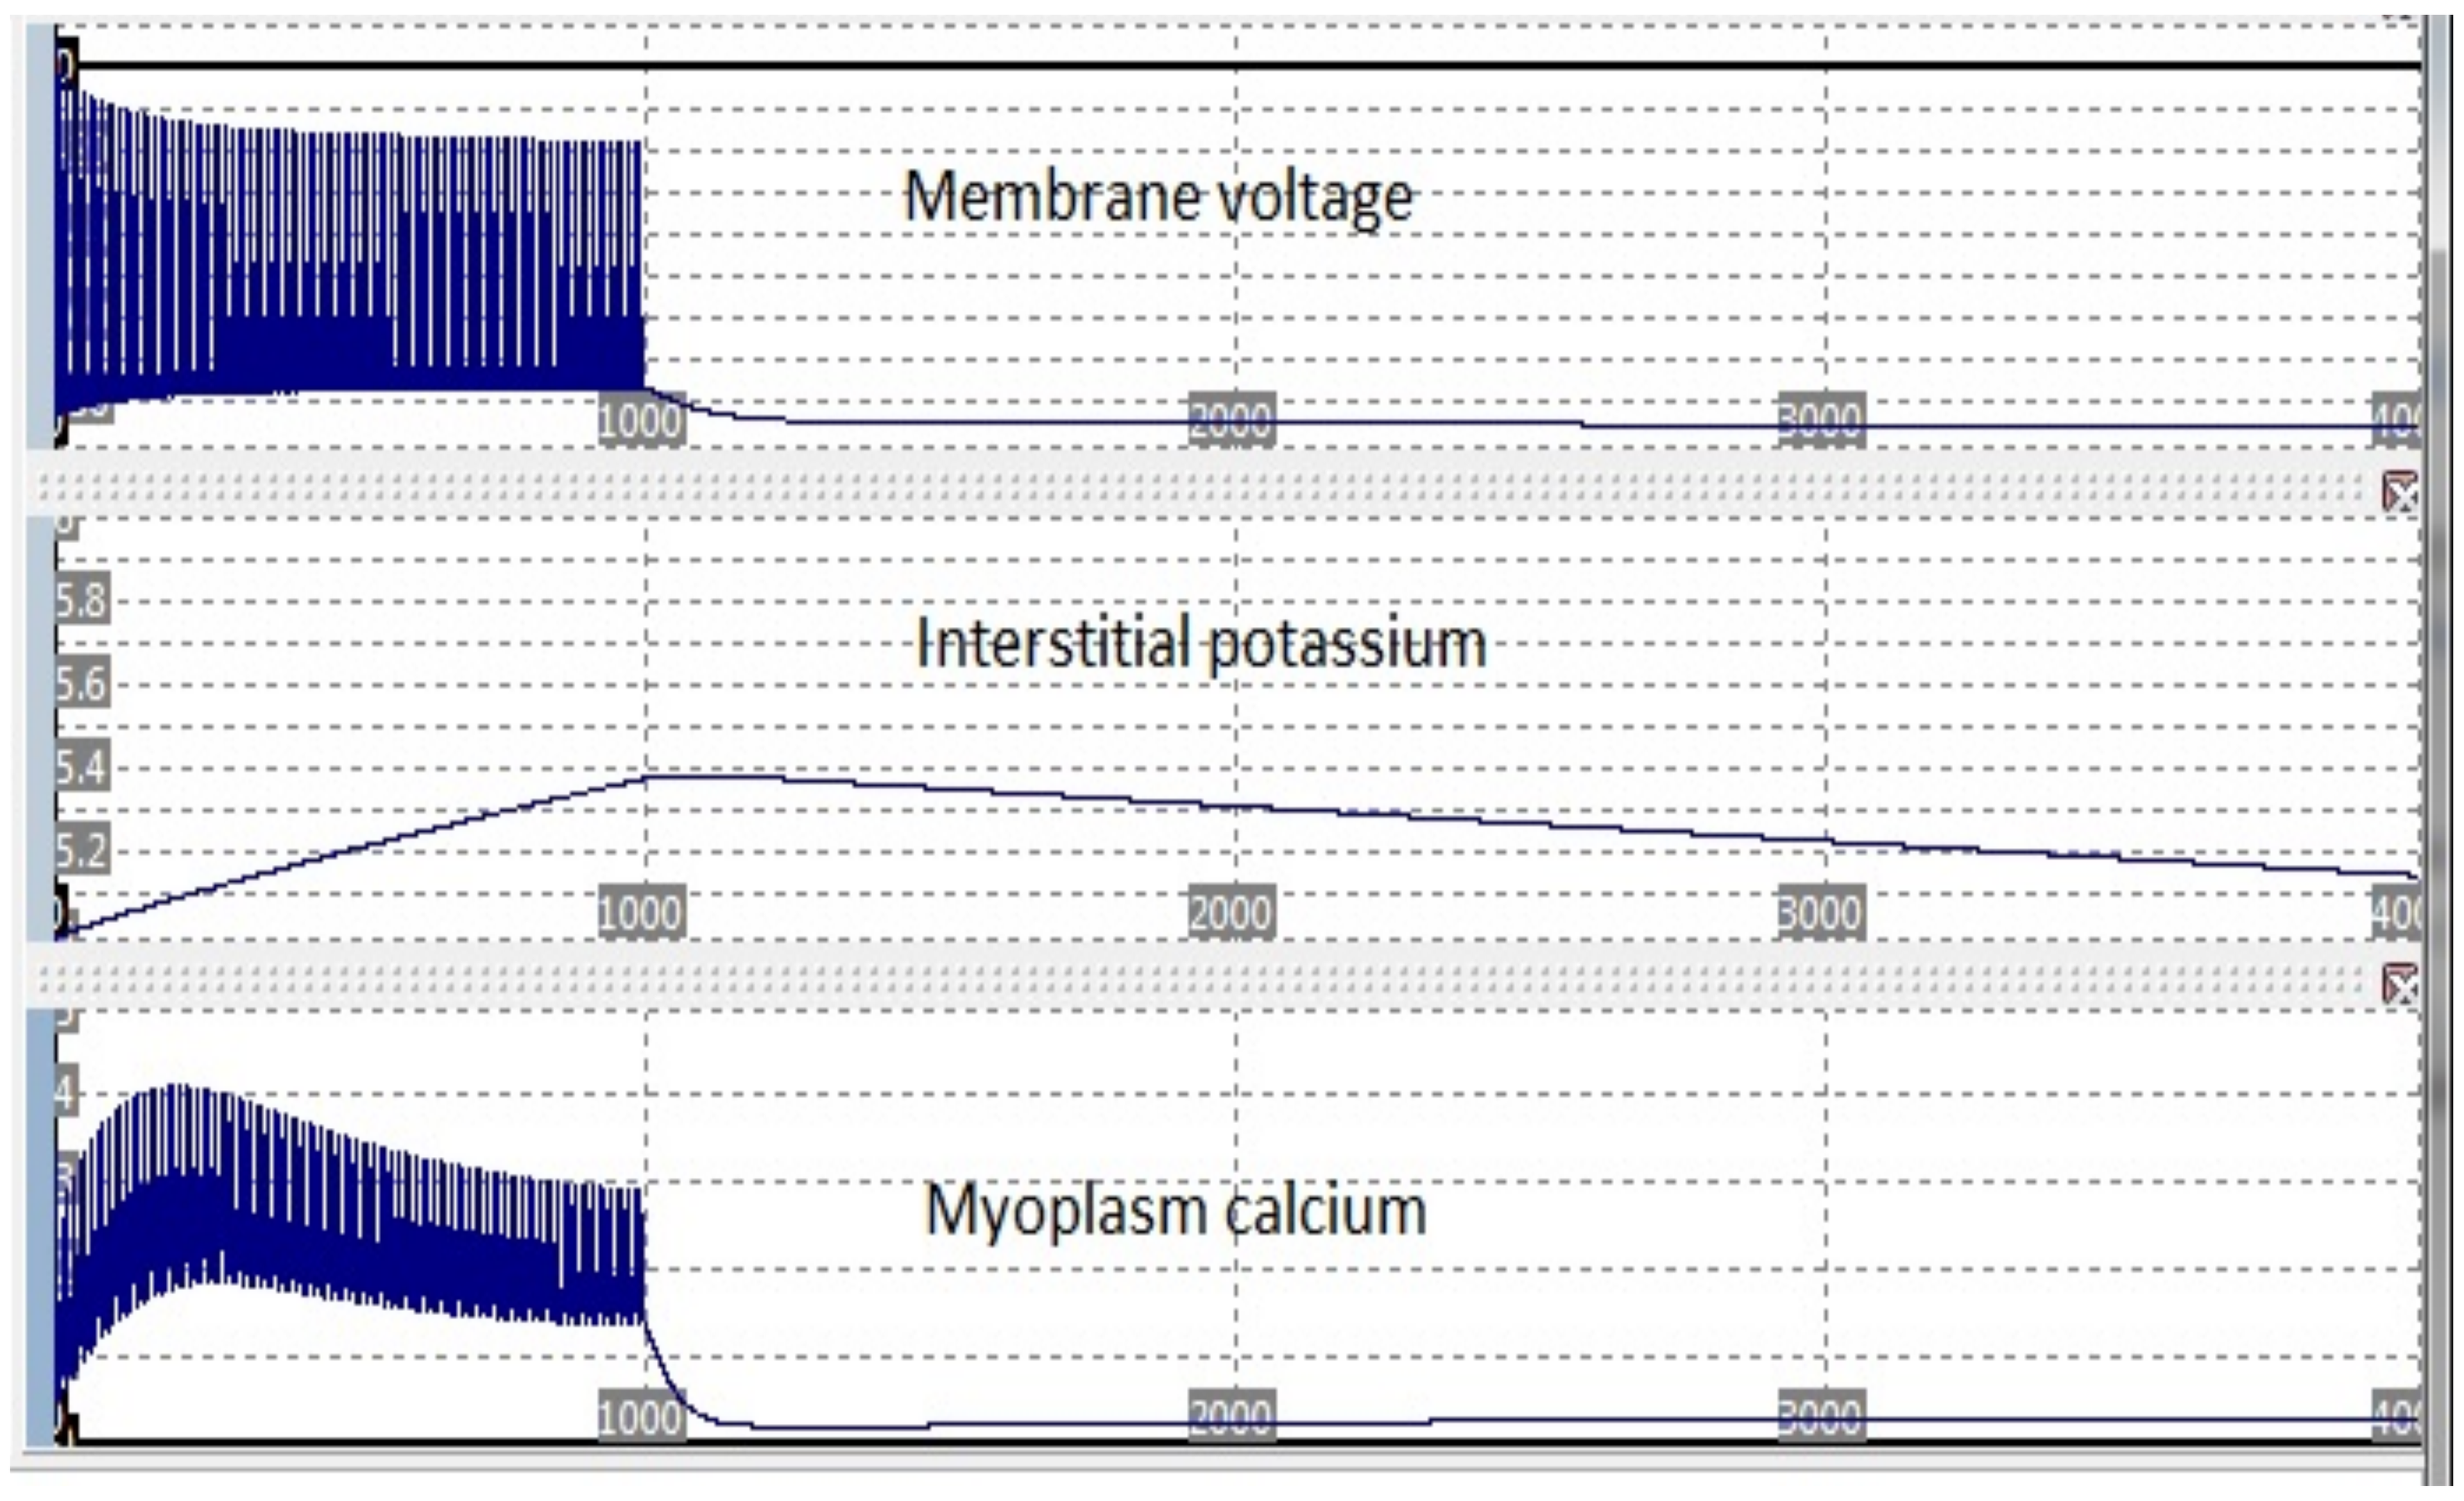
Membrane voltage
1000
2000
3000
Interstitial potassium
5.8
5.6
5.4
5.2
1000
2000
3000
Myoplasm calcium
1000
2000
3000