Effects of Glycyrrhetic Acid & Other Chemical Components of Shakuyakukanzoto on Cutaneous Microcirculation in Humans

Gabrielle A. Dillon[1], Sushant M. Ranadive[1], Michael J. Joyner[1], Andy R. Eugene[1], Kazuyo M. Tasaki[2], Denis Noble[2]
[1]Department of Anesthesiology, Mayo Clinic, Rochester, MN
[2]Department of Physiology, Anatomy and Genetics, University of Oxford, Oxford, UK

Abstract

Purpose: The purpose of this study was to explore dose dependent vasodilation responses of the individual compounds of Shakuyakukanzoto (SKT), a 1:1 combination of extracts from Paeony and Licorice roots: glycyrrhetic acid (GA), paeoniflorin (PF), and isoliquiritigenin (ISO).

Methods: 20 young, healthy participants (13M/7W) had four intradermal microdialysis fibers placed in the forearm skin for local delivery of: 1) control (diluent), 2) GA, 3) PF, or 4) ISO at doses of 1, 10, 50, or 100 µm. Cutaneous red cell flux, an index of skin blood flow, was measured by laser-Doppler flowmetry. Blood pressure (BP) was monitored continuously during fixed measurement periods via finger photo-plethsmography (Finapres). Cutaneous vascular conductance (CVC) was derived as red cell flux divided by BP and was normalized as a percentage of baseline (%CVCbl, saline at 4 µl/min) and the maximum CVC (%CVCmax, 28mM sodium nitroprusside at 4 µl/min, local temperature 43°C).

Results: Mean CVC did not statistically differ between baseline and individual compound plateau (average of last 5 minutes of infusion) at any dose. The peak flux was significantly higher from baseline at doses 50 and 100 µm for control (56.17±13.32 vs. 96.73±20.32 and 44.64±5.68 vs. 80.21±9.54), PF (62.70±10.09 vs. 115.72±16.16 and 62.37±16.2 vs. 150.21±34.26), GA (48.37±4.41 vs. 71.76±7.16 and 58.36±13.68 vs. 91.51±3.41), and ISO (96.50±23.20 vs. 128.57±27.90 and 44.95±8.78 vs. 91.18±20.37) ($p<0.05$). However, peak CVC was significantly higher only for GA at dose 50 µm, as well as for control and PF at dose 100 µm compared to their respected baselines (GA: 0.60±0.06 vs. 1.01±0.11, control: 0.61±0.09 vs.1.55±0.61, PF: 0.81±0.20 vs. 1.30±0.30, $p<0.05$). There were no significant differences between the control and the compounds for ΔCVC, %CVCbl, or %CVCmax. There were no significant differences between doses 1, 10, or 50 as compared to 100 µm for other variables.

Conclusion: These results suggest that the doses may be lower or below the threshold to cause a vasodilatory effect. Future studies that investigate higher doses of these compounds may be needed.

Background

- Shakuyakukanzoto (SKT) is an herbal preparation of the Paeony and Licorice roots, containing: glycyrrhetic acid (GA), paeoniflorin (PF), and isoliquiritigenin (ISO).
- SKT has been used for over 2,000 years as a dietary supplement in oriental medical practices. It has been used to treat muscle cramps in hemodialysis patients and duodenal spasms in peristalsis patients.[1, 2, 3]
- One potential mechanisms could involve the vasodilatory capacity of either one or all of the individuals compounds of SKT.
- However, it is currently unknown if any of the components of SKT namely GA, PF and ISO have vasodilatory potential.

Purpose

To explore dose dependent vasodilation responses of the individual compounds of Shakuyakukanzoto (SKT)

Methods

- 20 young, healthy participants (13M/7W) had four intradermal microdialysis fibers placed in the forearm skin for local delivery of: 1) control (diluent), 2) GA, 3) PF, or 4) ISO at doses of 1, 10, 50 or 100 µm.

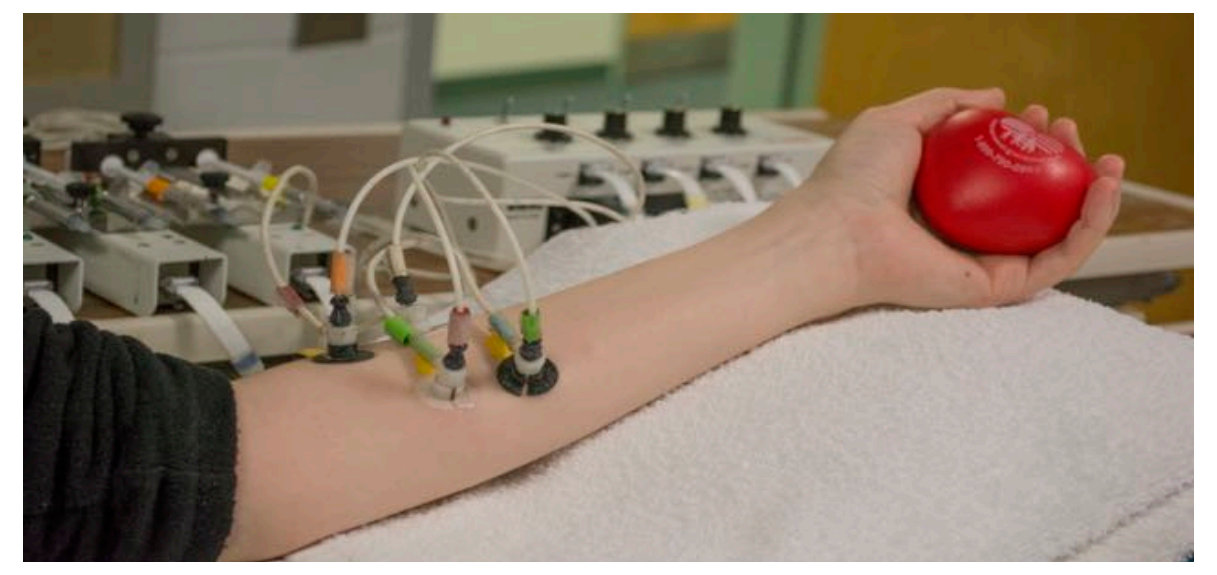

- Blood Pressure (BP) was monitored continuously during fixed measurement periods via finger photo-plethsmography (Finapres).
- Cutaneous red cell flux, an index of skin blood flow, was measured by laser-Doppler flowmetry.
- To normalize for BP, cutaneous vascular conductance was calculated as $CVC = (flux \cdot BP^{-1}) \cdot 100$.

Protocol:

- Baseline (saline) was infused for 1 hour for hyperemia to subside.
- Compounds were infused for 20 minutes; One compound was infused per site.
- Sodium nitroprusside was infused at a local heat of 43ºC for maximal dilation for about 30-40 minutes or until flow plateaued.

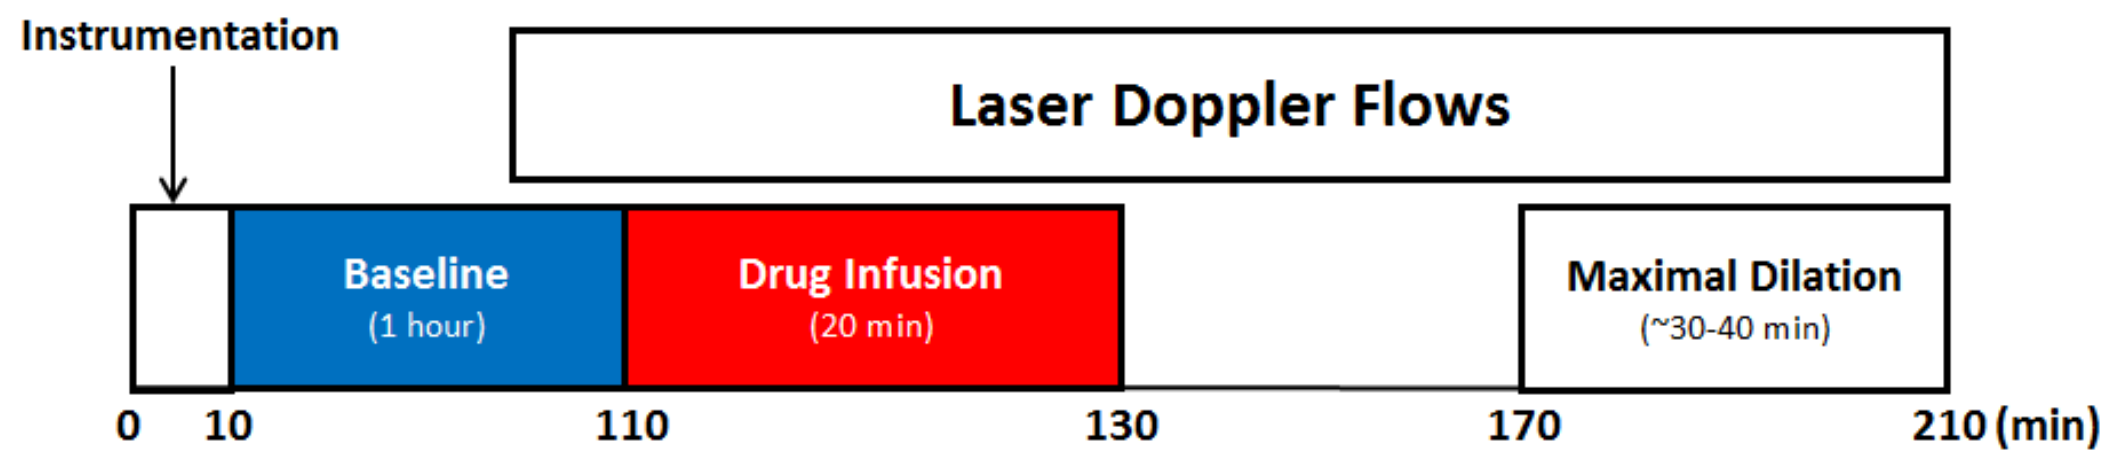
Instrumentation
Laser Doppler Flows
Baseline
(1 hour)
Drug Infusion
(20 min)
Maximal Dilation
(~30-40 min)
0
10
110
130
170
210 (min)

Figure 1. Timeline of protocol.

- Mean CVC was calculated as the average of the last five minutes of drug infusion.
- Peak CVC was defined as the highest CVC value taken from 15 second averages following the first minute of drug infusion.

Results

Variable	Dose 1µm (n=4)	Dose 10µm (n=4)	Dose 50µm (n=6)	Dose 100µm (n=6)
Age (yr)	31 ± 3	25 ± 2	30 ± 2	24 ± 2
Height (cm)	183 ± 3	177 ± 4	177 ± 6	178 ± 4
Weight (kg)	78 ± 4	73 ± 2	75 ± 3	70 ± 4
BMI (kg/m²)	23 ± 1	23 ± 1	24 ± 1	22 ± 1
MAP (mmHg)	90 ± 2	82 ± 5	82 ± 4	75 ± 3

Table 1. Participant characteristics for all groups showed similar physical and hemodynamic variables($p>0.05$). Values are means ± SE.

Drug	Dose 1µm (n=4)	Dose 10µm (n=4)	Dose 50µm (n=6)	Dose 100µm (n=6)
Control	0.88 ± 0.09	0.27 ± 0.18	0.09 ± 0.13	0.13 ± 0.08
GA	0.88 ± 0.09	0.17 ± 0.32	0.16 ± 0.10	0.05 ± 0.16
PF	0.14 ± 0.12	0.19 ± 0.15	0.11 ± 0.24	0.53 ± 0.33
ISO	-0.01 ± 0.06	0.06 ± 0.09	0.10 ± 0.13	0.12 ± 0.10

Table 2. ΔMean CVC for Control did not differ from any drug infusions at any dose($p>0.05$). Values are means ± SE.

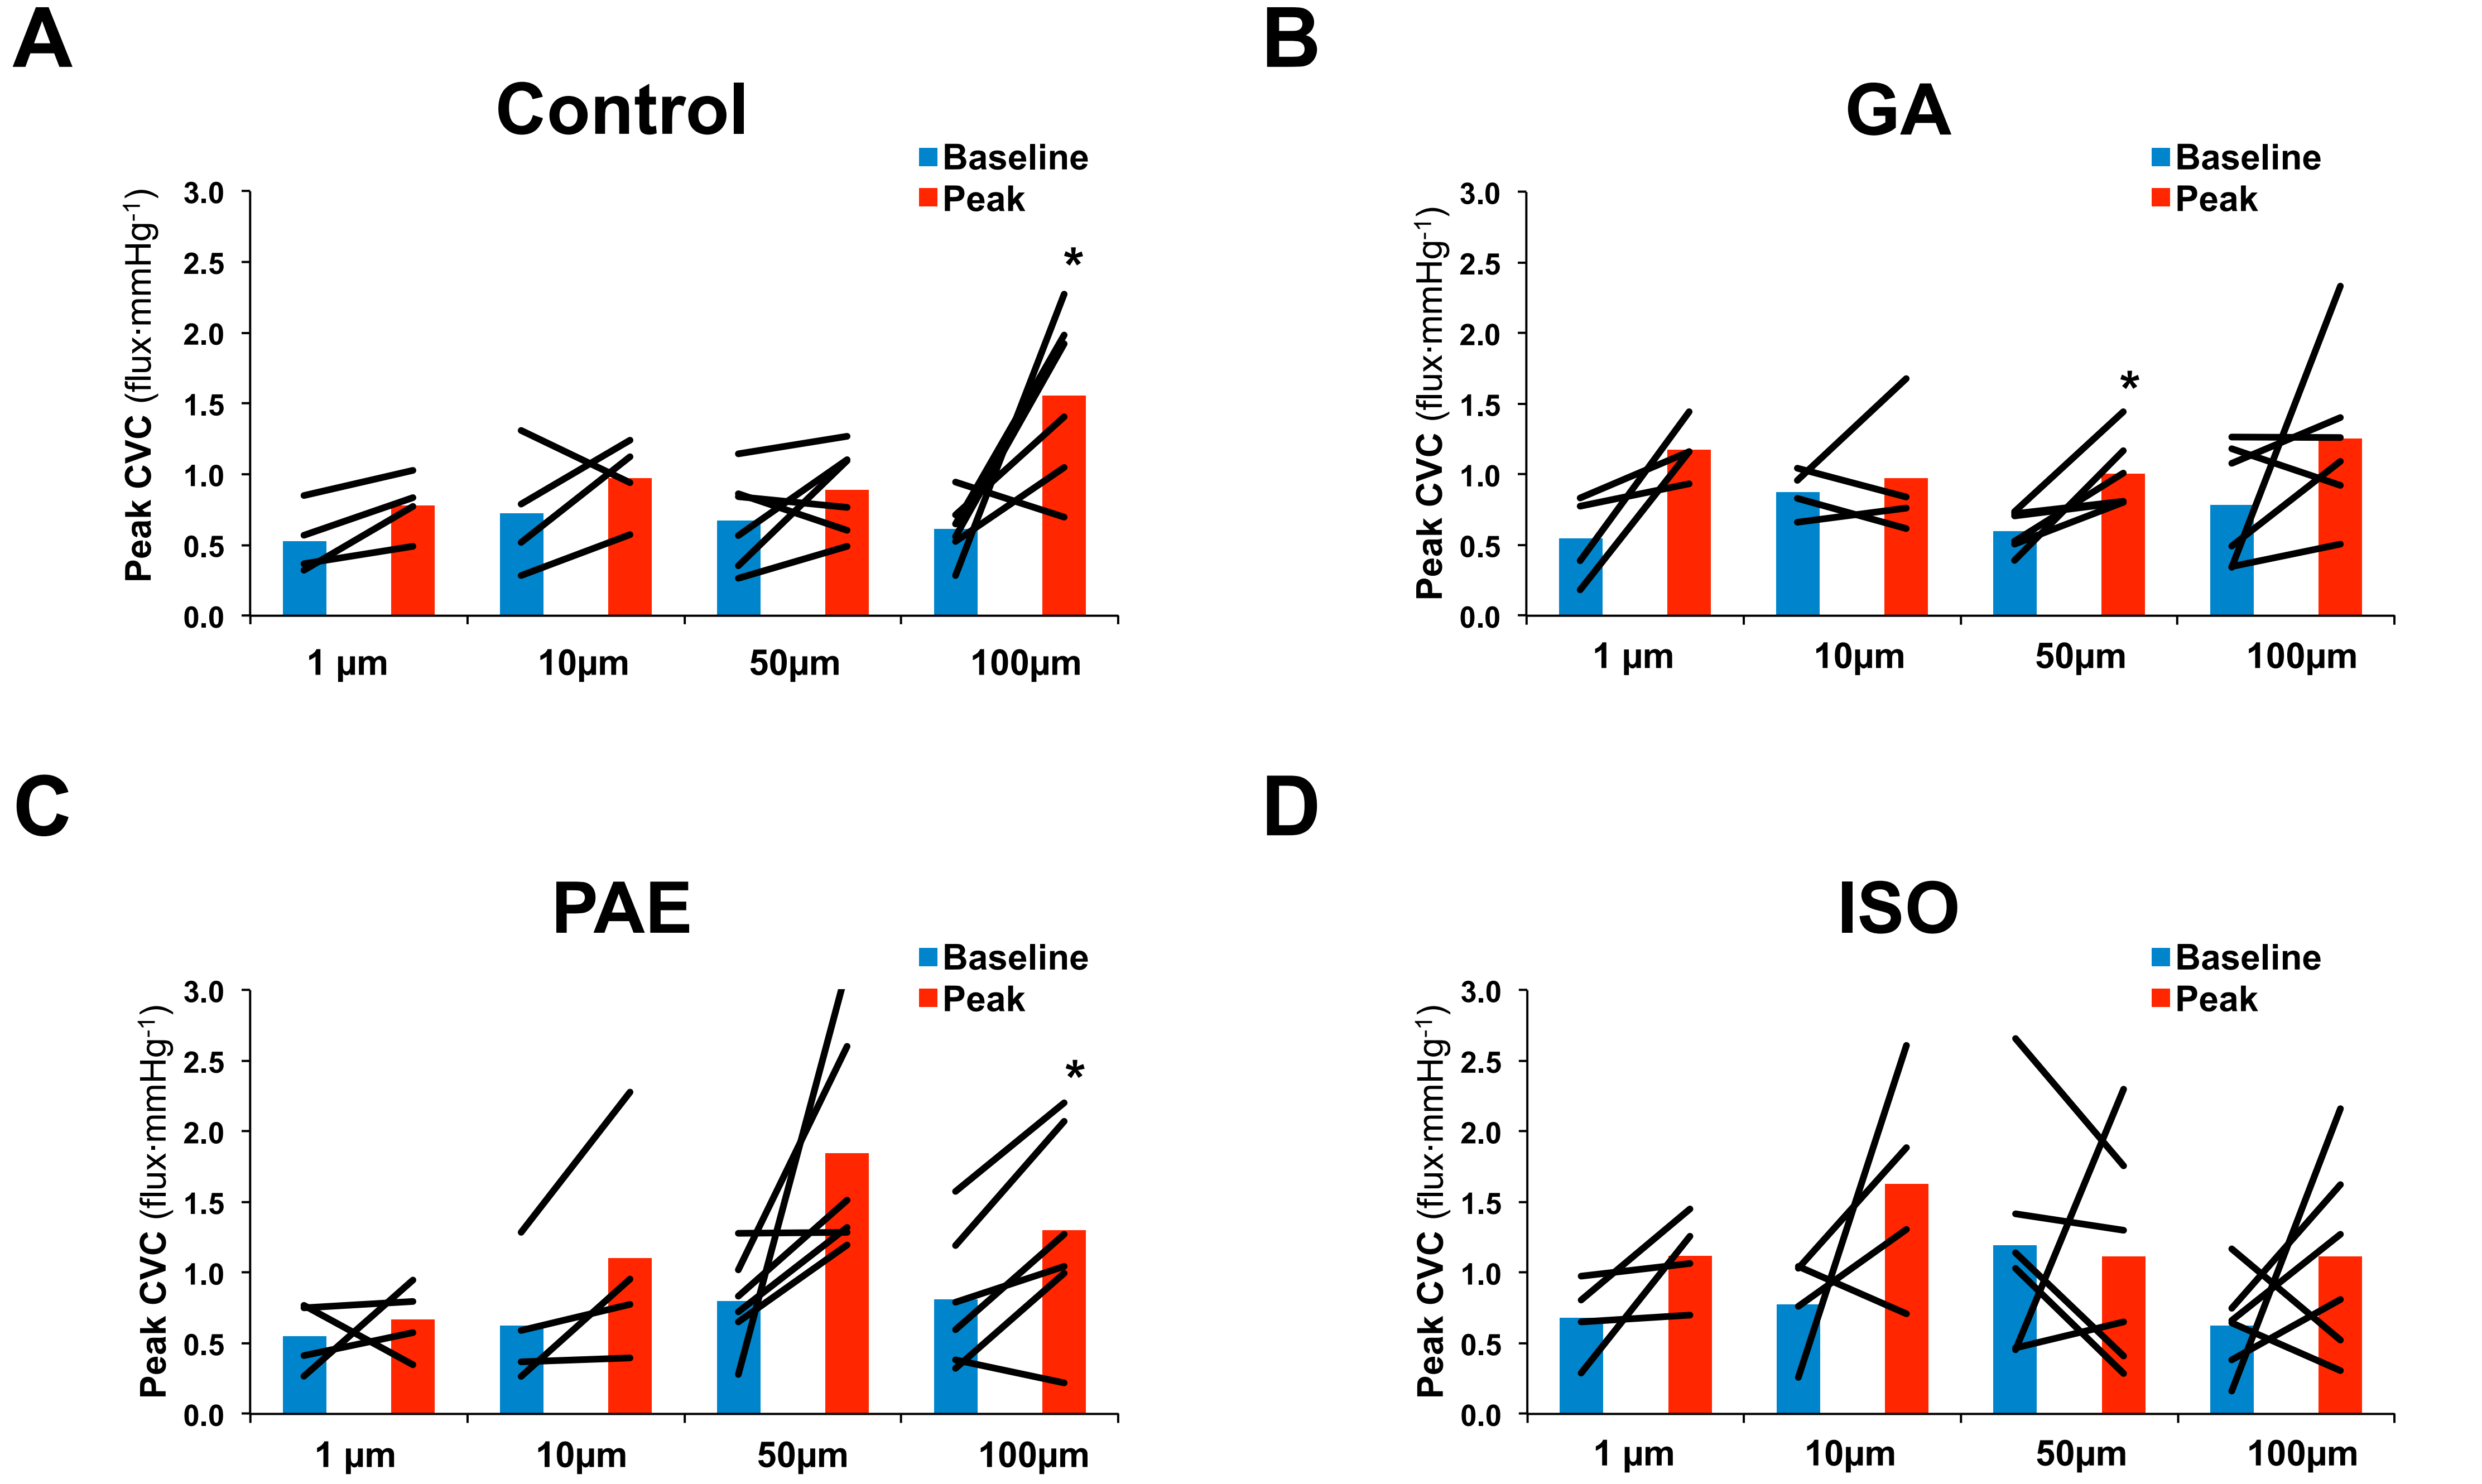
A
Control
B
GA
C
PAE
D
ISO
Baseline
Peak
Peak CVC (flux·mmHg⁻¹)
3.0
2.5
2.0
1.5
1.0
0.5
0.0
1 µm
10µm
50µm
100µm
*

Figure 2. Baseline and peak CVC responses to infusions of A) Control, B) GA, C) PAE, and D) ISO. Peak CVC for Control did not significantly differ from any drug infusions at any dose. *$p<0.05$ as compared to baseline.

Discussion

- Mean CVC did not statistically differ between baseline and individual compound plateau at any dose.
- There were no significant differences between the control and the compounds for ΔCVC, %CVCbl, or %CVCmax.
- There were no significant differences between doses 1, 10, or 50 as compared to 100 µm for ΔCVC, %CVCbl, or %CVCmax.
- Current doses did not cause significant vasodilation; higher doses should be investigated in future studies.
- Future studies could include investigating a synergistic effect of different compounds vs individual compounds and blocking individual vasodilatory pathways (i.e. NOS inhibition) may be needed.

Conclusions

These results suggest that the doses may be lower or below the threshold to cause a vasodilatory effect.

References and Acknowledgments

1. Hinoshita, F., Ogura, Y., Suzuki, Y., et al. Effect of orally administered shao-yao-gan-cao-tang (shakuyaku-kanzo-to) on muscle cramps in maintenance hemodialysis patients: a preliminary study. Am J Chin Med. 2003; 31 (3):445-53.
2. Hyodo, T., Taira, T., Takemura, T., et al. Immediate effect of Shakuyaku-kanzo-to on muscle cramp in hemodialysis patients. Nephron Clin Pract. 2006; 104 (1):c28-32.
3. Sakai, Y., Tsuyuguchi, T., Ishihara, T., et al. Confirmation of the antispasmodic effect of shakuyaku-kanzo-to (TJ-68), a Chinese herbal medicine, on the duodenal wall by direct spraying during endoscopic retrograde cholangiopancreatography. J Nat Med. 2009; 63 (2): 200-3.

Special Thanks to: our study volunteers and the Integrative Human Physiology Lab at St. Mary's Campus, Mayo Clinic.

Funding: The University of Oxford Systems Biology Project sponsored by Tsumura.

© 2016 Mayo Foundation for Medical Education and Research